My Life, Our Future: A multi-sector collaboration to provide genotyping services and a research repository for the hemophilia community expands from pilot to national program.

Diane Aschman, MS;[1] Marion Koerper, MD;[2] Sarah Ruuska, MPH;[3] Sally McAlister, RN, BSN;[4] Elaine Eyster, MD, FACP;[5] Lisa Baker, RN, BSN;[5] Michael Tarantino, MD;[6] Sarah Gonzales, MT (ASCP), CCRP;[6] Barbara Konkle, MD[3]

[1] American Thrombosis and Hemostasis Network, Chicago, IL; [2] University of California, San Francisco, San Francisco, CA; [3] Puget Sound Blood Center, Seattle, WA; [4] Biogen Idec, Weston, MA; [5] Penn State School of Medicine, Hershey, PA; [6] Bleeding and Clotting Disorders Institute, Peoria, IL

OBJECTIVES

Hemophilia A and B are lifelong, X-linked recessive disorders that result in a deficiency of clotting factor VIII or factor IX, respectively.[1] There are approximately 16,000 individuals with hemophilia A and 4,000 with hemophilia B in the U.S.[2] Approximately 70% of all people with hemophilia have an inherited gene mutation, while the remaining 30% of cases are caused by a spontaneous mutation.[3] Hemophilia genotype provides meaningful information about bleeding severity, inhibitor risk, carrier detection and prenatal diagnosis. With only 20% of U.S. patients with hemophilia having received genotype analysis, the U.S. lags behind other developed nations. A multi-sector collaboration, known as *My Life, Our Future* (MLOF), was formed to launch a program addressing this need, while also establishing a repository of associated samples and data to support future scientific discovery and treatment advances in hemophilia.

METHODOLOGY

MLOF offers free genotype analysis to patients with hemophilia A or B, and has created a research repository of blood samples and genetic data that could be matched to phenotypic data collected separately. The program is a formal collaboration among four healthcare entities, with representation on a steering committee that guides the program:

- American Thrombosis and Hemostasis Network (ATHN) – working with 135+ affiliated hemophilia treatment centers (HTCs), provides HTC provider education and secure infrastructure for data collection
- National Hemophilia Foundation (NHF) – with 52 chapters, educates consumers and supports recruitment
- Puget Sound Blood Center (PSBC) – central genotyping laboratory and sample repository
- Biogen Idec – scientific collaboration and initiative support

Combining their expertise with the HTCs that deliver genetic services, the partners implemented a pilot program, including a suite of educational materials to support each step of the process with patients and HTCs. *(Table 1)*

Over ☞

Table 1. Tools to Support Program Implementation

Process Goal	Support Mechanisms
Patient awareness	NHF Annual Meeting presentations, brochure, video, exhibit; *HemAware* article; local NHF chapter presentations; www.MyLifeOurFuture.org (includes FAQ, resource center, list of participating HTCs)
HTC recruitment	ATHN Data Summit presentations, breakout groups, exhibit; interest form and participation agreement; ATHNreport articles; start-up webinars; Biogen Idec Medical Science Liaisons
Patient enrollment	IRB-approved recruitment materials (brochure, fliers, letters to patients, phone script); HTC contact with patients during comprehensive care visits
HTC implementation support	HTC implementation webinars, PSBC 1-on-1 clinical research associate support, PSBC lab interpretation report, ATHN Clinical Manager system technical assistance
Research repository	PSBC sample storage, ATHN data stewardship
Dissemination of findings	Posters at professional meetings (e.g., APHA, APHL), ATHN research review process, presentations at community events (e.g., chapter annual meetings)

Figure 1. Cumulative Patient Enrollments By Month

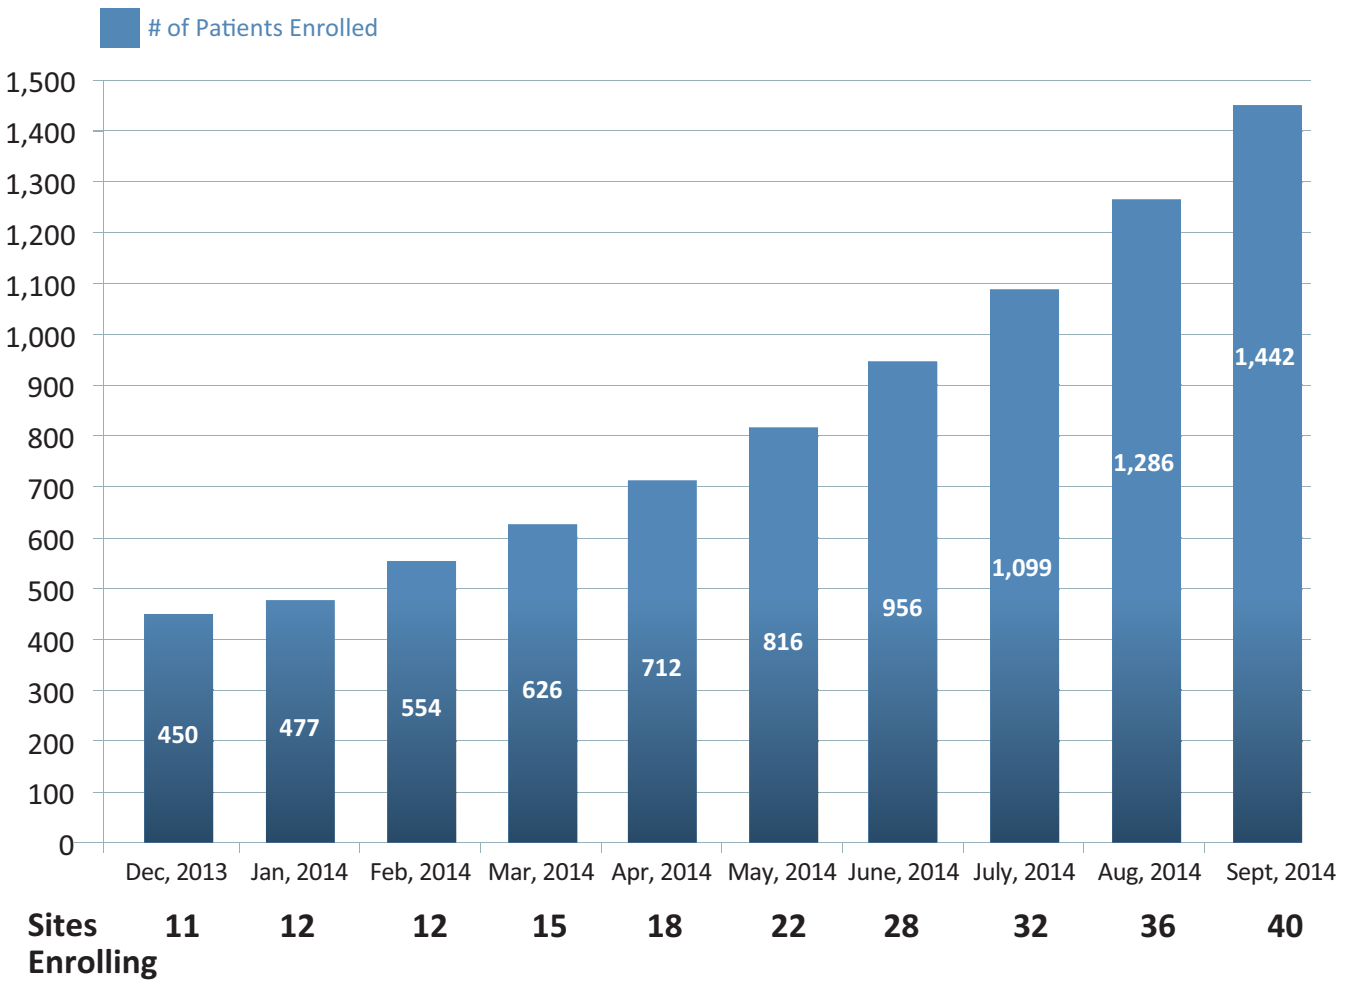

of Patients Enrolled
1,500
1,400
1,300
1,200
1,100
1,000
900
800
700
600
500
400
300
200
100
0
450
477
554
626
712
816
956
1,099
1,286
1,442
Dec, 2013
Jan, 2014
Feb, 2014
Mar, 2014
Apr, 2014
May, 2014
June, 2014
July, 2014
Aug, 2014
Sept, 2014
Sites Enrolling
11
12
12
15
18
22
28
32
36
40

RESULTS

Following the successful pilot with 11 HTCs, MLOF was open to all 137. As of September 30, 2014:

- 40 HTCs have enrolled 1,442 patients into the project. *(Figure 1)*
- After the initial start-up phase, HTCs have recruited an average of 4-5 patients per HTC per month
- Batch processing of the samples is done in increments of 192, so reports are typically returned to HTCs in 2-3 months. To date, clinical genotyping reports have been sent to providers for 952 patients (66% of samples collected)
- For patients who consent to contribute to research, genetic mutation findings are deposited in the data repository and samples are stored for future scientific study
- 81% of total enrolled patients consented to research (1,168 repository patients)

This project can serve as a model for providing genetic information to patients and creating a resource for scientific investigation in other rare diseases.

Of the 1,442 patients who received genotyping through one of 40 HTCs, 1,151 (80%) had hemophilia A and 291 (20%) had hemophilia B. Demographic breakdown by sex and hemophilia severity is shown in *Table 2*. The vast majority of those tested were male and about half had severe disease.

Table 2. Patient Demographics by Hemophilia Type, Severity and Sex

Hemophilia Type	Males			Females			Total
	Severe	Moderate	Mild	Severe	Moderate	Mild	
A	602	210	296	2	6	35	1151
B	103	112	68	0	0	8	291
Total	705	322	364	2	6	43	1442

Most frequent mutation type was missense: 46% in hemophilia A, 77% in hemophilia B. In patients with severe hemophilia A, 42% were due to inversion 22 or 1 mutations. By comparison to available hemophilia A and B databases, 109 mutations not previously reported were identified in 114 patients. *(Figure 2)*

Figure 2. Newly Identified Mutations by Mutation Type (n=114 patients)

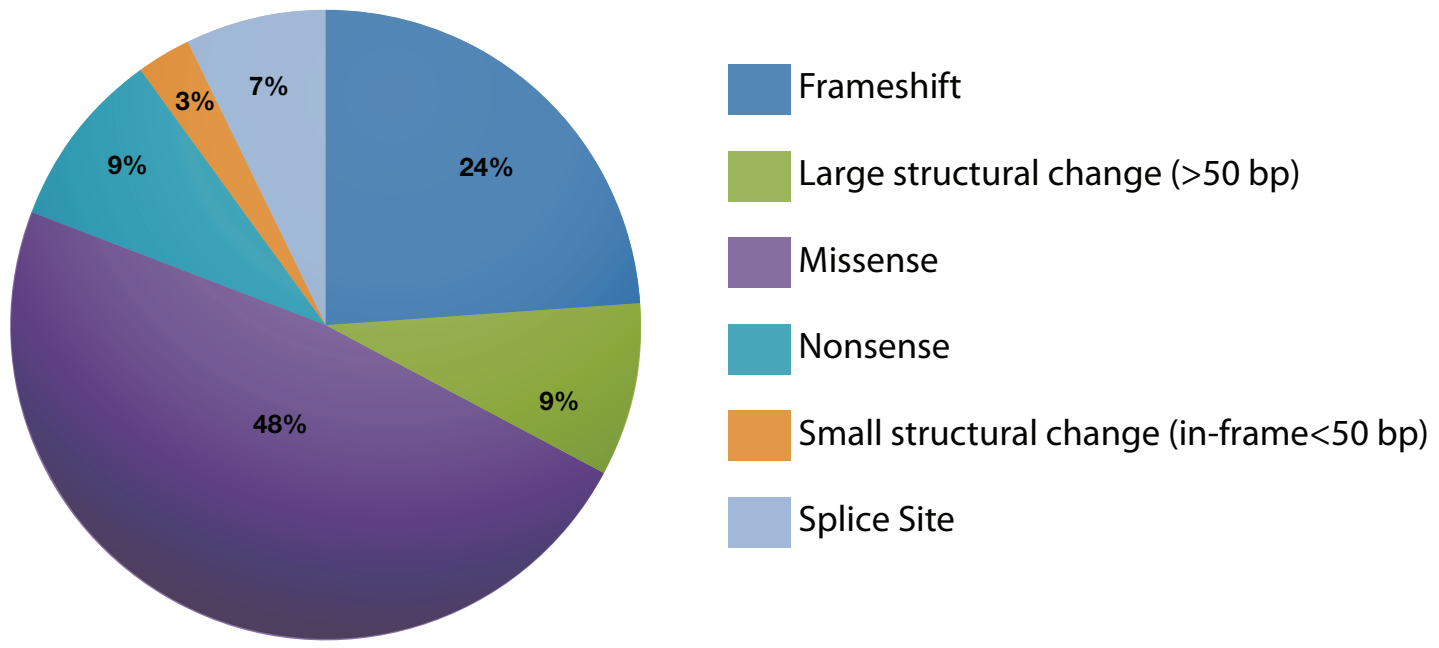

24%
9%
48%
9%
3%
7%
Frameshift
Large structural change (>50 bp)
Missense
Nonsense
Small structural change (in-frame<50 bp)
Splice Site

The overwhelming majority of patients learned of the program through direct contact with their HTC or MLOF educational programs. Priority questions addressed through the educational programs included patient privacy protections, staff requirements, amount of blood required and system-related issues. Following the initial implementation period, participating sites recommended additional tools to assist HTCs in documenting familial relationships with patients, establishing pedigrees for carriers and educating participants about their mutation when results are returned.

CONCLUSIONS

Through collaboration, the partners are providing all U.S. hemophilia A and B patients access to free genotype testing through participating HTCs, and building a data and bio-repository to support future research. This project can inform other rare disorders by providing a model for the collection of genetic information to further research.

REFERENCES

[1] Mannucci, P and Tuddenham, EGD; The Hemophilias – from Royal Genes to Gene3 Therapy; N Engl J Med, 2001;344: 1773-1779.

[2] CDC. Hemophilia: Data & Statistics in the United States, updated May 7, 2013. Available at http://www.cdc.gov/ncbddd/hemophilia/data.html. Accessed July 9, 2014.

[3] National Hemophilia Foundation. Hemophilia A. https://www.hemophilia.org/Bleeding Disorders/Types-of-Bleeding-Disorders/Hemophilia-A. Accessed October 9, 2014.

American Public Health Association's Annual Meeting & Exposition – November 15-19, 2014

mylife ourfuture
Genotyping for Progress in Hemophilia